PRINT Particles Enable Liquid Storage of Incompatible Components: A case study with HibTT and DTaP

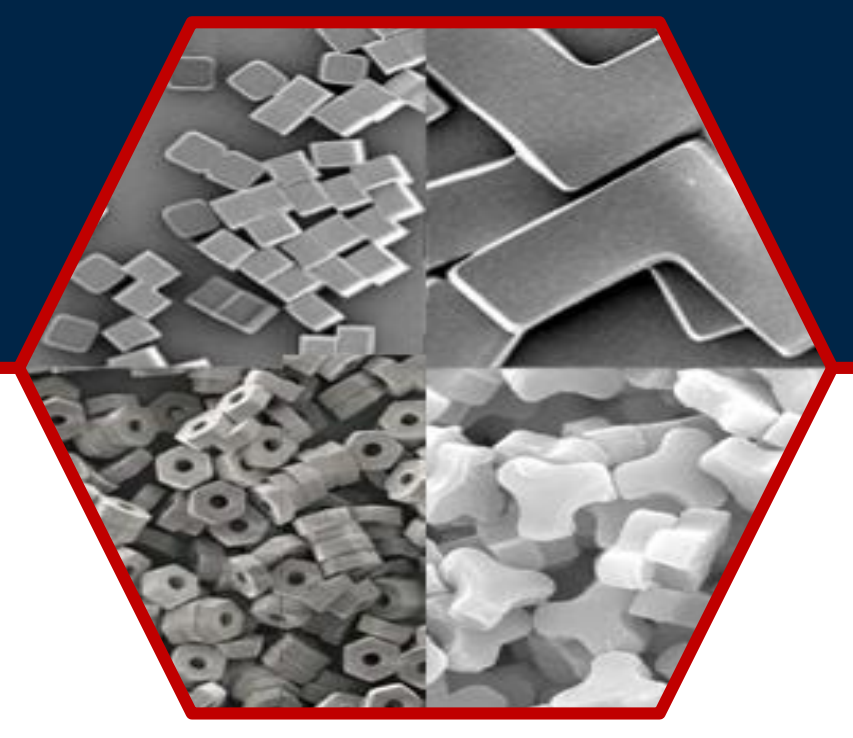

Ashley Galloway[1*], Laurent Strodiot[2*], Nadia Ouaked[2], Jin Lee[1], Patrick Pohlhaus[1], Ramya Yadavalli[1], Gabe Fawcett[1], Meredith Earl[1], Joseph Marchand[1], Marie-Noëlle Donner[2], Stéphane Temmerman[2], Nathalie Garçon[2], Eric De Buyl[2], Fabrice Godfroid[2], Abdelatif Elouahabi[2*], and Michele Stone[1*]

[1] Liquidia Technologies, Inc. Research Triangle Park, NC USA, [2] GSK Vaccines, Rixensart, Belgium, * Authors contributed equally to this work.

Introduction

When Hib-TT, *Haemophilus influenzae* type b tetanus toxoid conjugate vaccine, is combined with pediatric DTaP vaccines, a reduction in Hib antibody titer has been reported in infants as compared to the Hib-TT standalone vaccine.[1,2,3] DTaP vaccines contain aluminum hydroxide which is known to increase the immune response to diphtheria, tetanus and pertussis antigens[4]. However, Hib polysaccharides irreversibly bind on aluminum hydroxide crystals resulting in the depolymerization of Hib polysaccharides[5] which leads to a reduction in antigen availability and immunogenicity[3].

Herein we report a successful approach to segregate and protect Hib-TT from degradation catalyzed by aluminum hydroxide during storage, while maintaining strong immune responses in rats post immunization to Hib-TT as well as to DTaP antigens co-stored with Hib-TT-particles.

Liquidia's PRINT Process

PRINT, an acronym for Particle Replication In Non-wetting Templates, is a technology that allows precise control of particle design features such size, shape, and composition. The PRINT technology is uniquely suited to mold particles out of a variety of materials.[6-9]

The core process involves four basic steps:

1. Create a film of the desired composition on a delivery sheet.
2. Laminate the film with a mold where the material fills the mold cavities.
3. Remove particles from the mold.
4. Collect particles to create a particle suspension or dry powder.

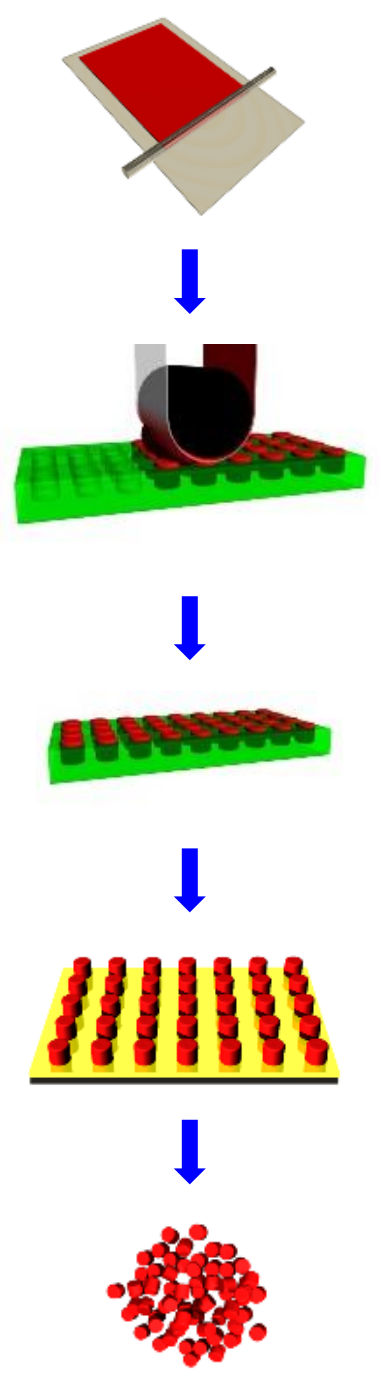

There are several variables that can be leveraged to create particles of a wide range of shapes, sizes, and chemical and physical composition.

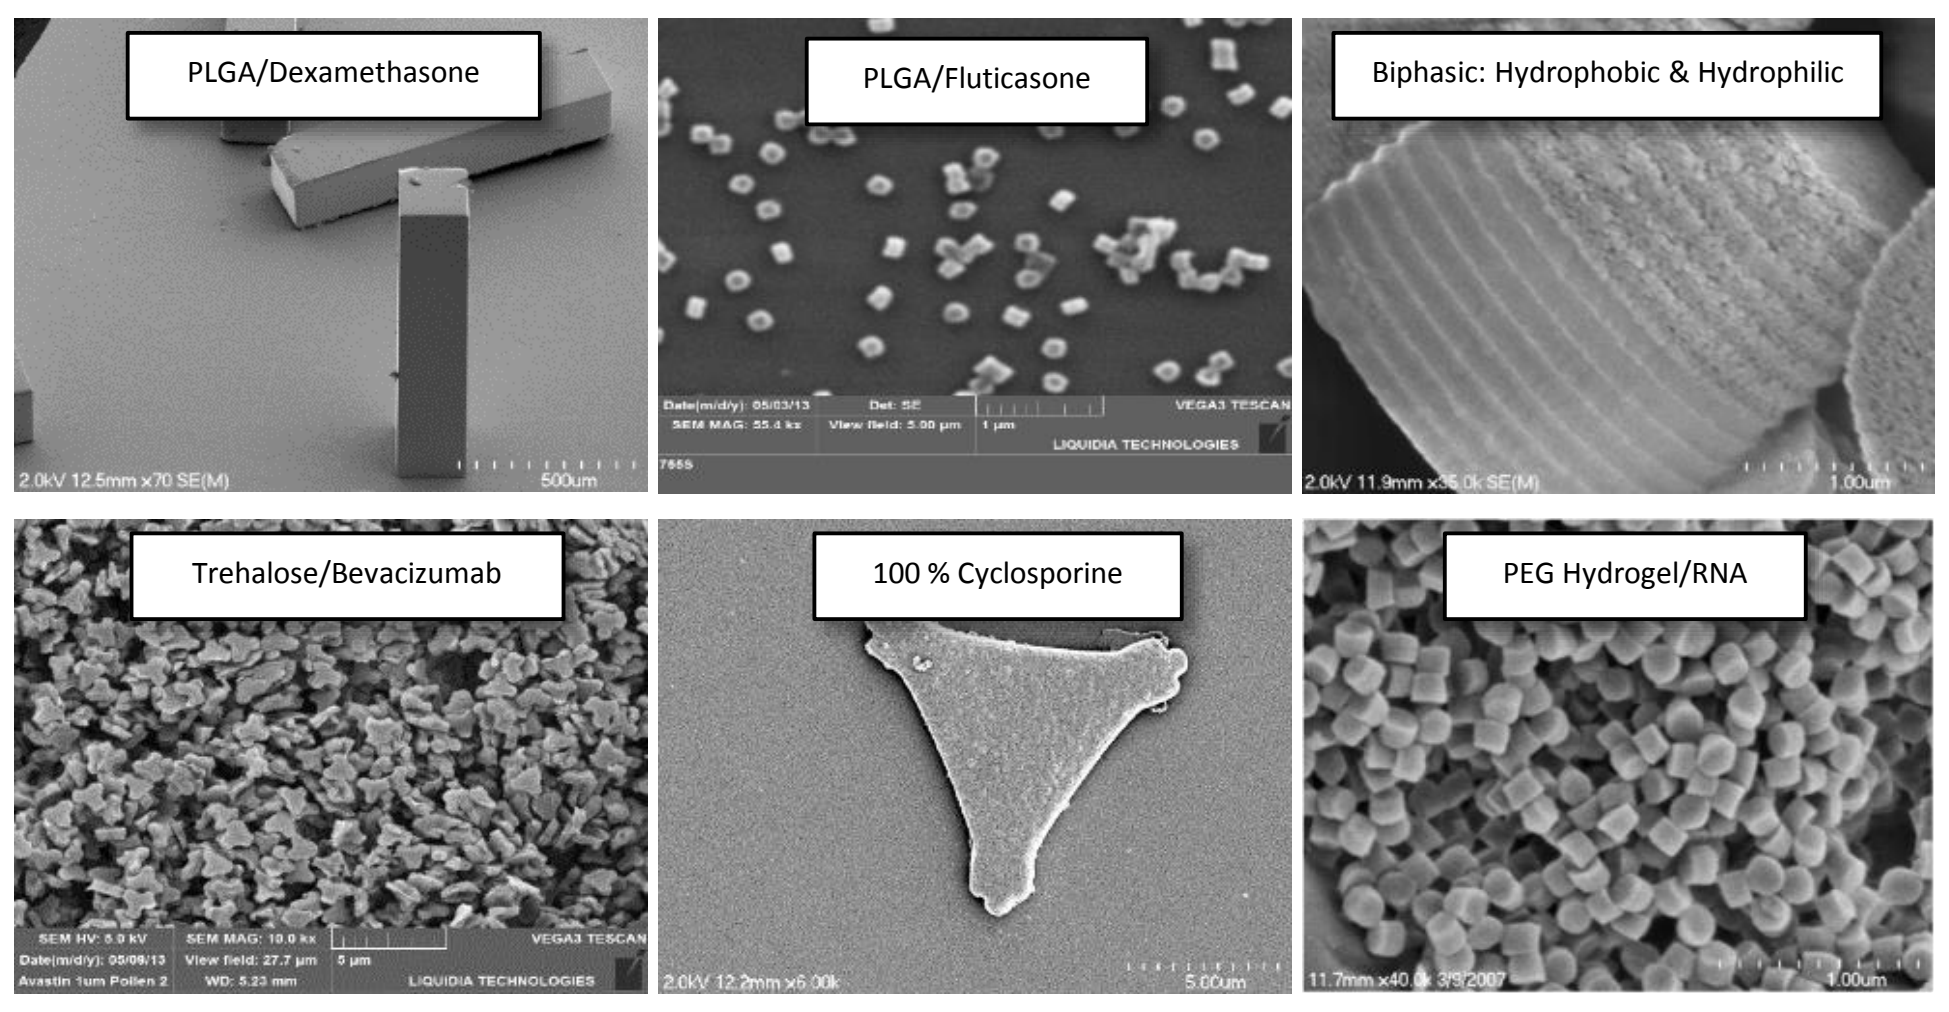
PLGA/Dexamethasone
PLGA/Fluticasone
Biphasic: Hydrophobic & Hydrophilic
Trehalose/Bevacizumab
100 % Cyclosporine
PEG Hydrogel/RNA

Materials

When trying to segregate Hib-TT in particles, it was desirable to find a material with low solubility at the desired storage conditions for the vaccine and a high solubility at physiological conditions after intramuscular injection. For this purpose, a 2:1 copolymer of poly(methyl methacrylate-co-methacrylic acid) commercially available as Eudragit S100 (ES100) from Evonik Industries, was selected.

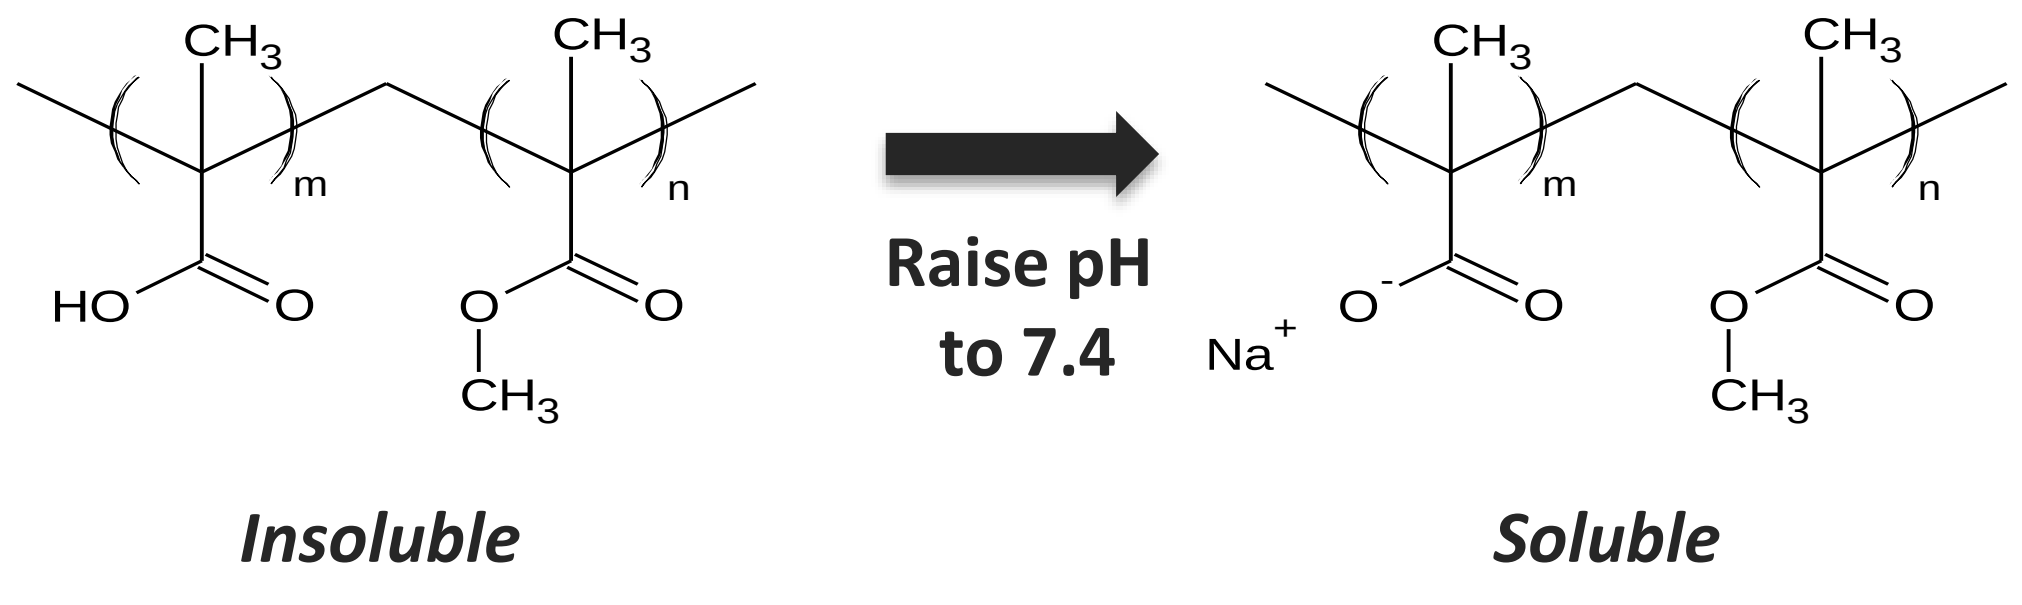
Raise pH to 7.4
Insoluble
Soluble

Particle Design

Using Liquidia's PRINT process, 6 μm donut shaped microparticles were prepared from ES100 material blended with Hib-TT.

Optical image of ES100/Hib-TT particles

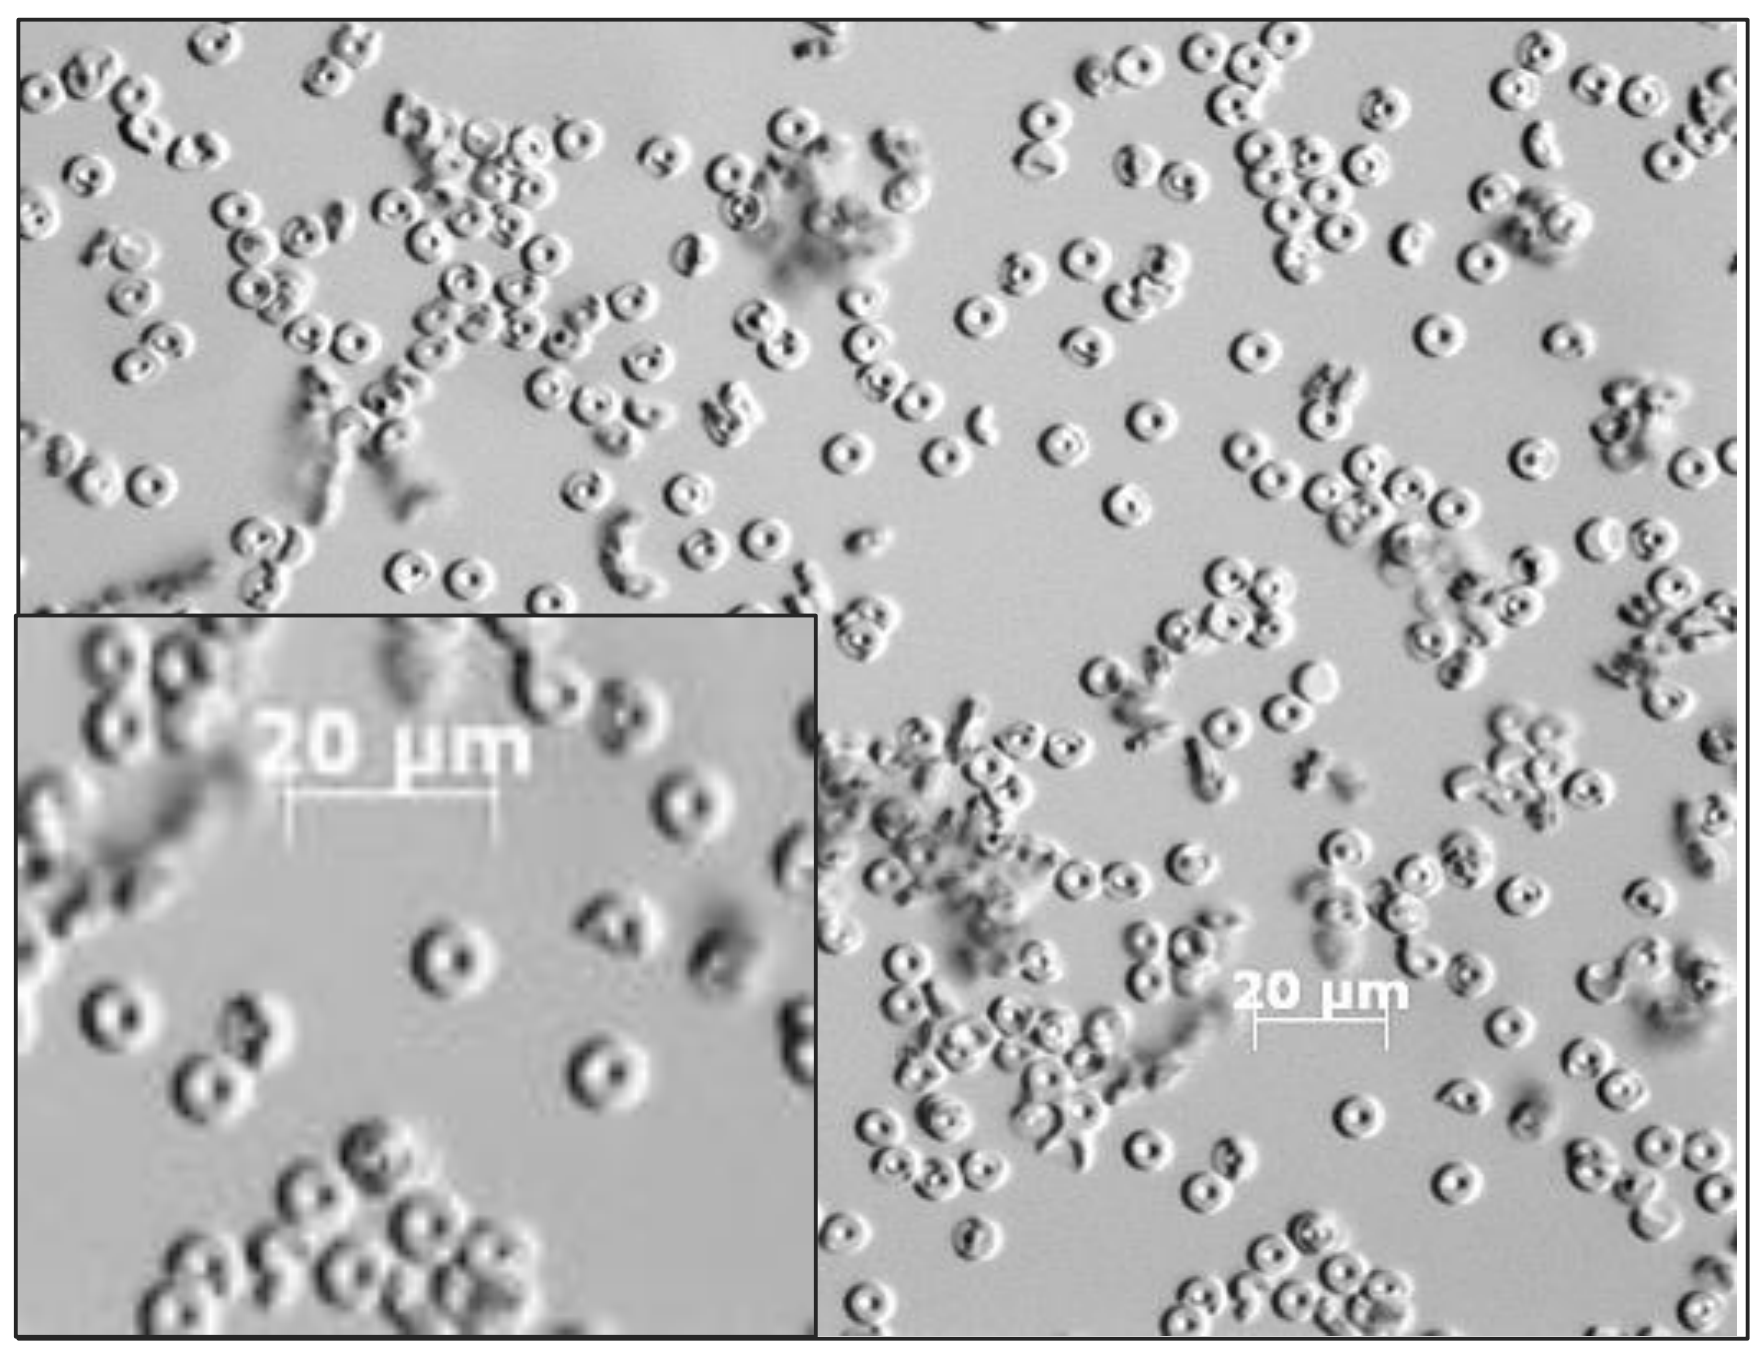
20 μm
20 μm

Particles remain insoluble at pH 6.1 and dissolve upon raising the pH above 7.

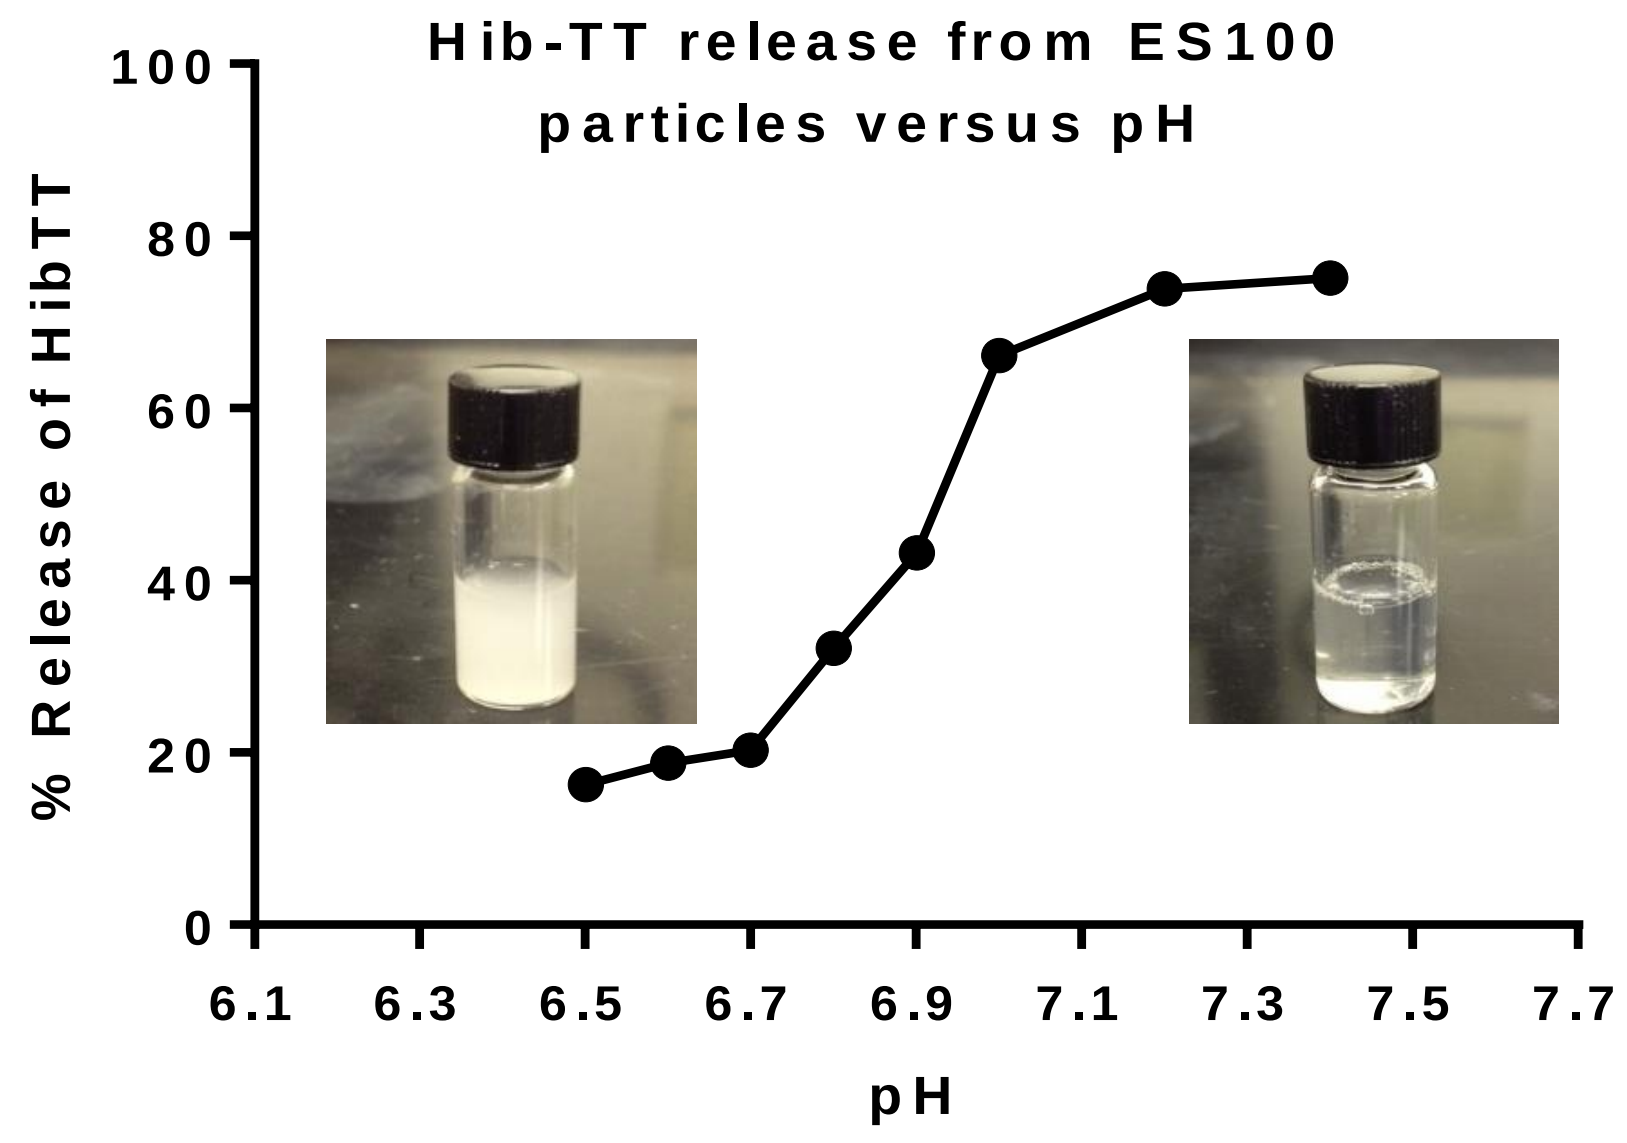
Hib-TT release from ES100 particles versus pH
% Release of HibTT
pH
0
20
40
60
80
100
6.1
6.3
6.5
6.7
6.9
7.1
7.3
7.5
7.7

Particle Characterization & Stability

After PRINT particles were formed, they were suspended in a suitable buffer and mixed with aluminum hydroxide. Mixed product was kept at 2-8°C and studied over time using several techniques including enzyme-linked immunosorbent assay (ELISA) to demonstrate antigen stability and antigenicity. Importantly, a blocking step was added to the ELISA to maintain antigen availability by preventing Hib-TT adsorption onto $Al(OH)_3$ once Hib-TT is released from particles into solution for measurement.

ELISA data indicated that Hib remained antigenic after storage with aluminum hydroxide for at least 16 months, demonstrating that the particles protected Hib-TT from the deleterious effects of storage with $Al(OH)_3$.

Protection of encapsulated Hib-TT from Alum adsorption

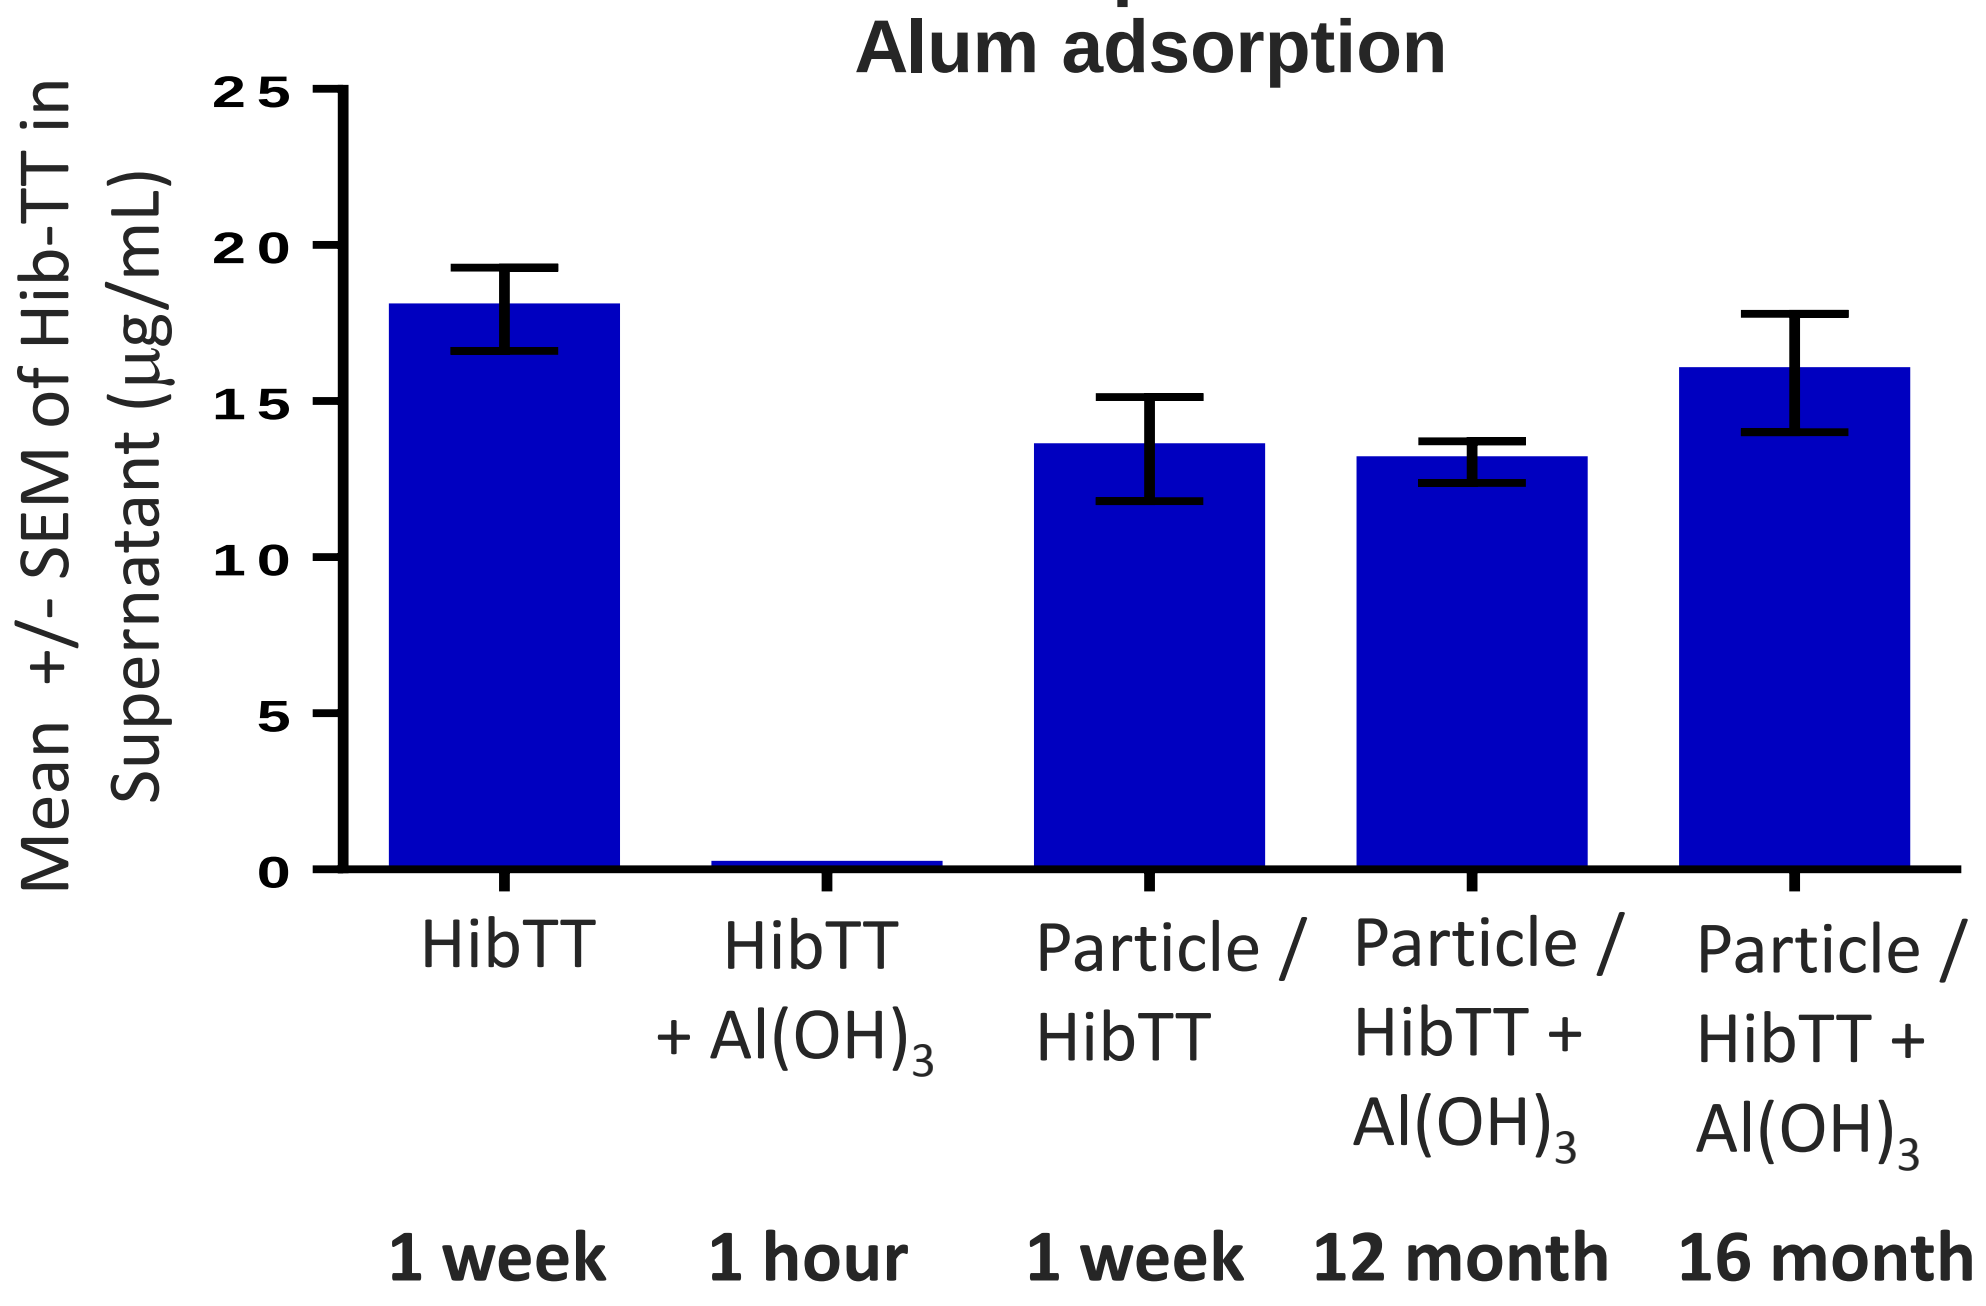
Mean +/- SEM of Hib-TT in Supernatant (μg/mL)
0
5
10
15
20
25
HibTT
HibTT + Al(OH)3
Particle / HibTT
Particle / HibTT + Al(OH)3
Particle / HibTT + Al(OH)3
1 week
1 hour
1 week
12 month
16 month

Immunogenicity Response

Three IM injections of PRINT particles containing 1 μg Hib-TT stored with $Al(OH)_3$ or as separate injections were administered in 6 week old adult female OFA rats (n=20). Hib-TT containing particles produced similar Hib antibody titers after the third injection whether or not the particles were stored with $Al(OH)_3$, indicating that the particles had protected Hib-TT. A similar response was observed for Hib-TT containing particles that had been stored with $Al(OH)_3$ for sixteen months, further demonstrating long term stability. Diphtheria, Tetanus, and Pertussis antigens that were co-formulated with the $Al(OH)_3$ and vialed with the Hib-TT containing particles also maintained equivalent immunogenicity to benchmark formulation (not shown).

7dPIII Serology anti-Hib PRR'P

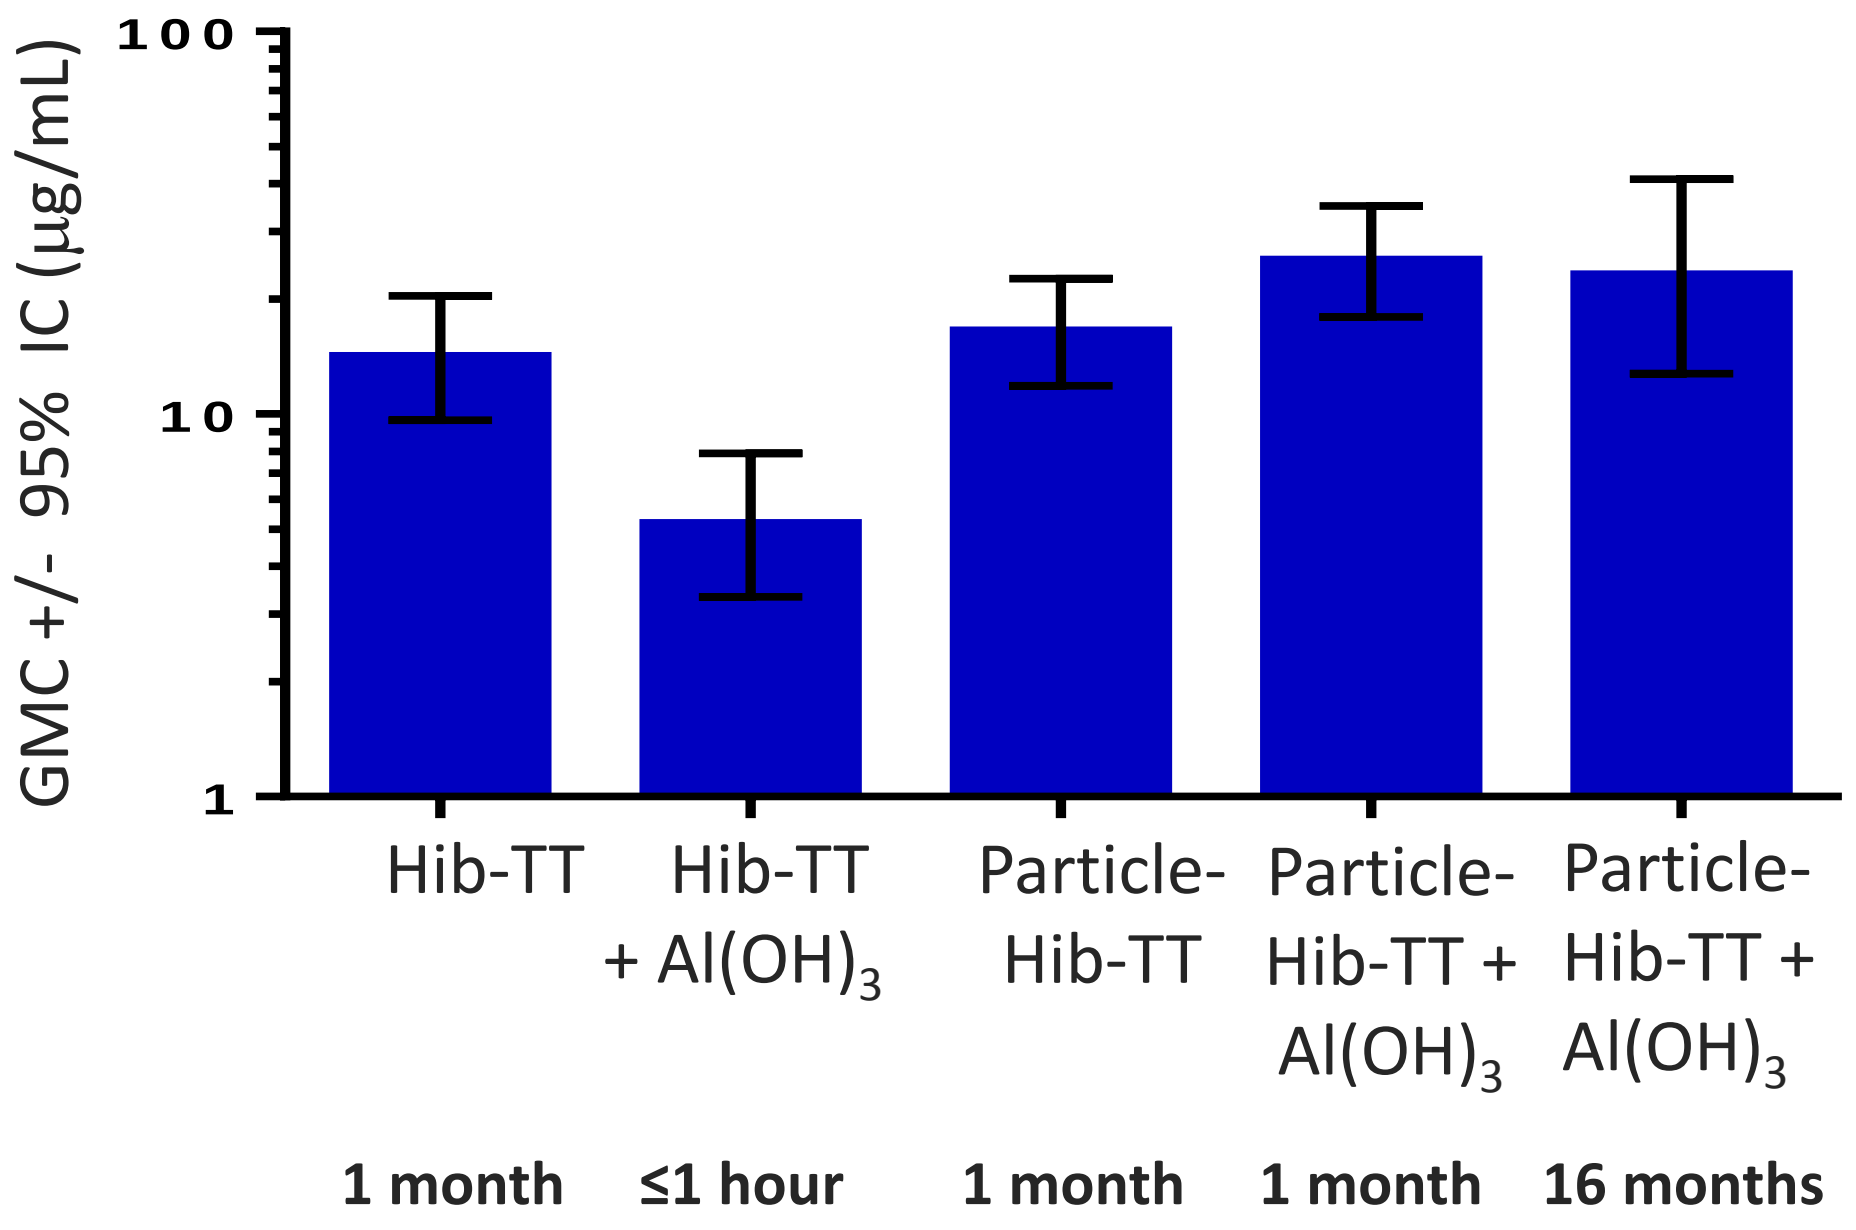
GMC +/- 95% IC (μg/mL)
1
10
100
Hib-TT
Hib-TT + Al(OH)3
Particle-Hib-TT
Particle-Hib-TT + Al(OH)3
Particle-Hib-TT + Al(OH)3
1 month
≤1 hour
1 month
1 month
16 months

Conclusions

PRINT particles prepared from ES100 material, a 2:1 copolymer of poly(methyl methacrylate-co-methacrylic acid), protected Hib-TT from degradation for at least 16 months as evidenced by Hib-TT integrity and Hib-immunogenicity data in a rat model while having no impact on the immune response to other antigens in the vaccine. This data establishes proof of concept that ES100-based PRINT particles can protect an antigen during storage with incompatible components and release that antigen upon dose administration to achieve a desired immunological response. Local and systemic studies in rats show no adverse signs of short-term reactogenicity under these experimental conditions (data not shown). Further studies are warranted to understand the ES100 based PRINT particle system and its applicability to aid in formulation of other vaccines or drug products.

Conflict of Interest Statement

This work was performed under a vaccines collaboration and option agreement between GlaxoSmithKline Biologicals SA and Liquidia Technologies Inc. Both companies were involved in all stages of the study conduct and analysis. The study was funded by GlaxoSmithKline Biologicals SA. Conflict of interest: LS, NO, MND, ST, NG, EDB, FG, and AE are, or were at the time of the study, employees of the GSK group of companies. AG, JL, PP, RY, GF, ME, LM, and MS are, or were at the time of the study, employees of Liquidia Technologies Inc.

References

1. White C. *Expert Rev Vaccines.* **2009**, *8*, 831–40
2. Skibinski DAG. *J Glob Infect Dis.* **2011**, *3(1)*, 63-72.
3. Mawas F. *Vaccine* **2006**, *24(17)*, 3505-12.
4. Otto RBD. *Biologicals* **2015**, *43*, 355-362.
5. Sturgess AW. *Vaccine* **1999**, *17*, 1169-78.
6. Galloway, AL. *Nanomedicine: NBM* **2013**, *9(4)*, 523–531.
7. Rolland, JP. *J. Am. Chem. Soc*. **2005**, *127(28)*, 10096–10100.
8. Kelly, JY. *J. Am. Chem. Soc.* **2008**, *130(16)*, 5438–5439.
9. Enlow, EM. *Nano Letters*. **2011**, 11(2), 808–813.

LIQUIDIA TECHNOLOGIES

gsk